Evaluation of *nab*-Sirolimus in Combination With PI3K Pathway Inhibitors to Overcome PI3K/mTOR Resistance in PI3K-Mutant Breast Cancer Cell Lines

A117

Sean Wallace, PhD[1]; Khine Nyein Myint, PhD[1]; Shihe Hou, PhD[2]; Maria Zalath, BA[2]; Andrew Kwon, PhD[2]; Brian McMorran, PhD[2]; Igor Vivanco, PhD[1]

[1]Institute of Pharmaceutical Science, King's College London, London, UK; [2]Aadi Bioscience, Pacific Palisades, CA, USA

Objective

- **To determine if combining *nab*-sirolimus with other PI3K/AKT/mTOR pathway inhibitors impacts cytotoxicity of anticancer agents in PI3K-mutant breast cancer cells**

KEY FINDINGS

The antitumor effects of PI3K and AKT inhibitors were enhanced by the addition of the novel mTOR inhibitor *nab*-sirolimus; the PI3K + mTOR inhibitor combination in particular was synergistic

The improved effectiveness of the PI3K and mTOR inhibitor combination may be due to overcoming resistance mechanisms induced by single-agent treatment

The combination of an mTOR ATP competitive inhibitor + allosteric inhibitor enhanced inhibition of the downstream mTOR target, phospho-4EBP1

These data support a vertical PI3K pathway inhibition strategy using *nab*-sirolimus and PI3K or AKT inhibitors in *PIK3CA*-mutated breast cancer, regardless of hormone receptor status

Presented at the AACR-NCI-EORTC International Conference on Molecular Targets and Cancer Therapeutics; Boston, MA; October 11-15, 2023

BACKGROUND

- The PI3K-AKT-mTOR pathway is often overactivated in many cancer types and plays a central role in chemotherapy resistance in solid tumors[1] (**Figure 1**)

Figure 1. PI3K-AKT-mTOR pathway

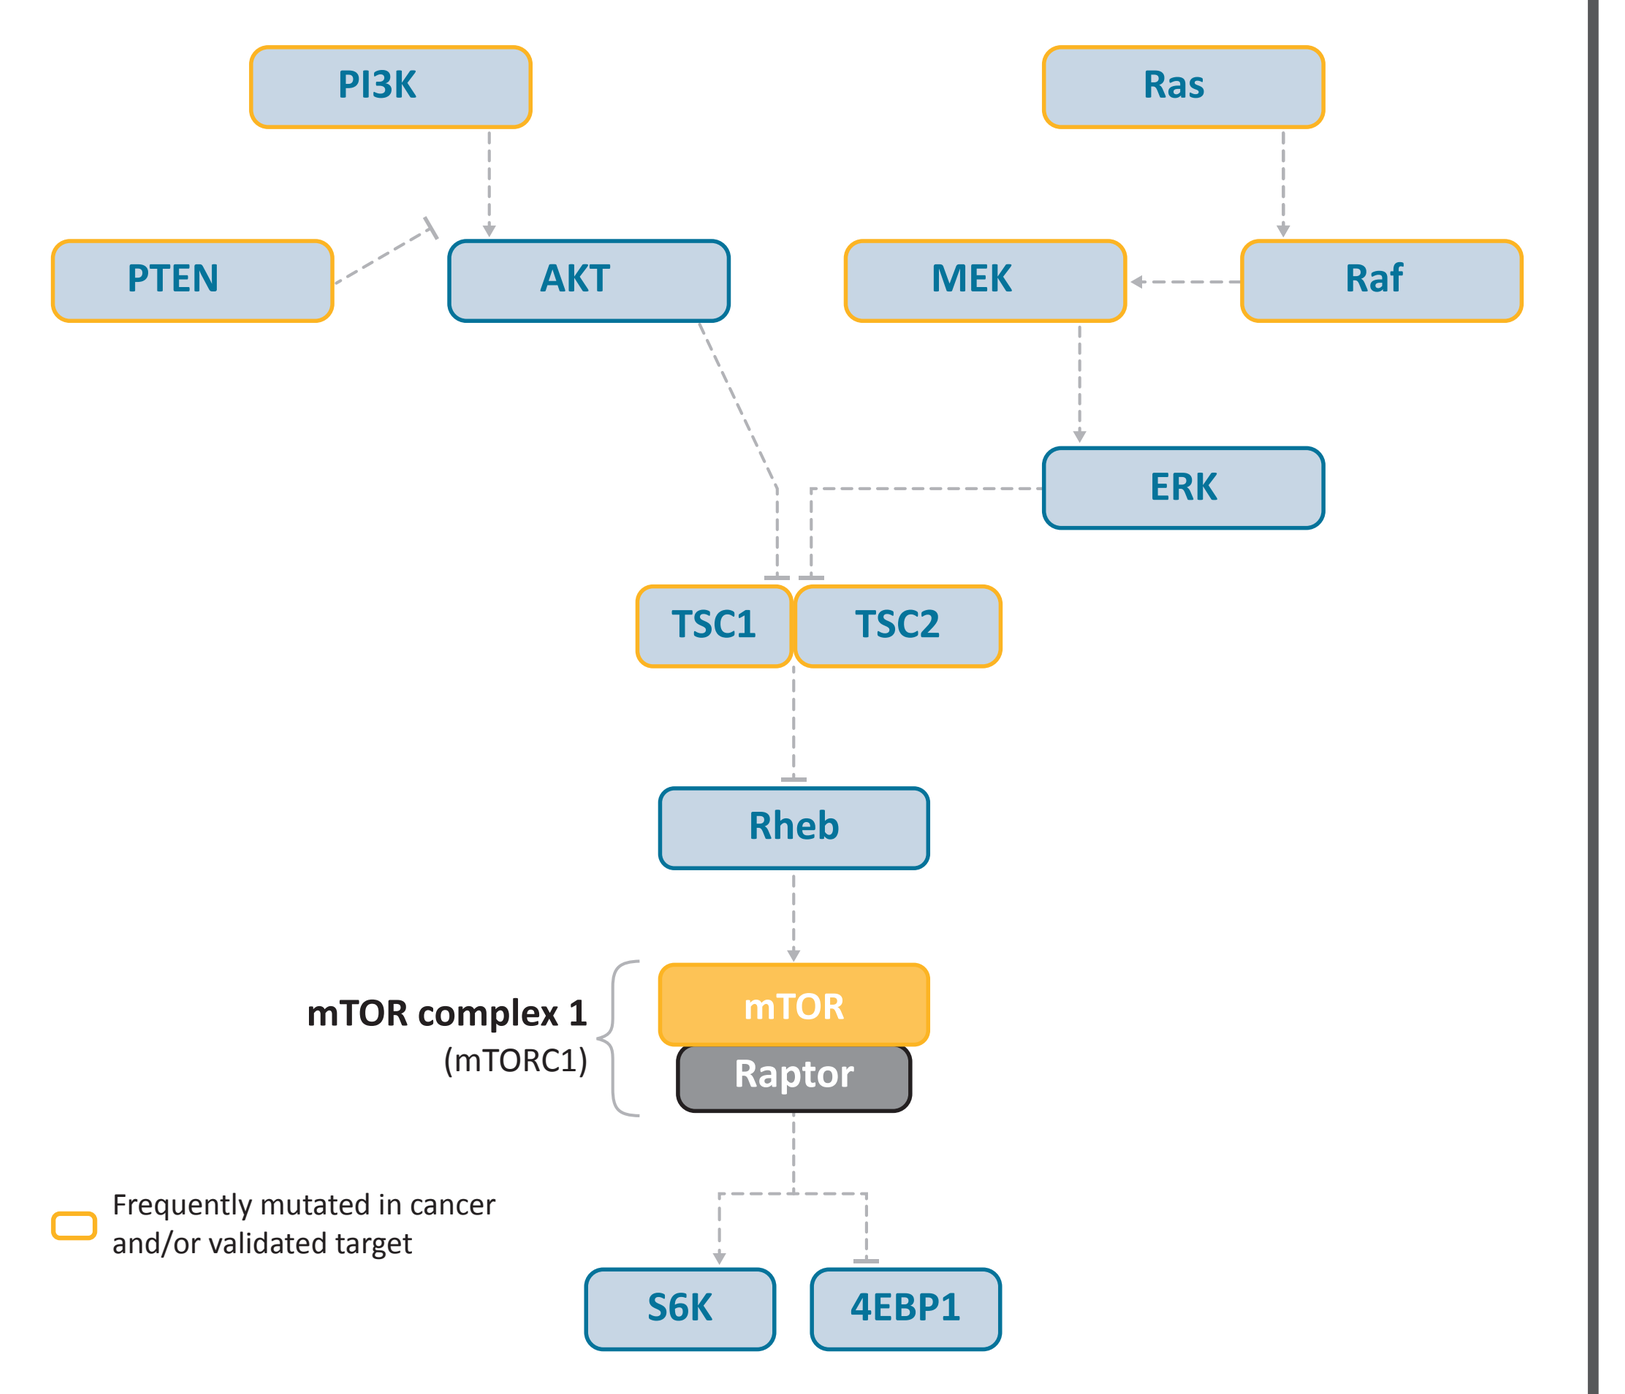

PI3K
Ras
PTEN
AKT
MEK
Raf
ERK
TSC1
TSC2
Rheb
mTOR complex 1
(mTORC1)
mTOR
Raptor
Frequently mutated in cancer and/or validated target
S6K
4EBP1

4EBP, eukaryotic translation initiation factor 4E-binding protein; **AKT**, protein kinase B; **ERK**, extracellular signal-regulated kinase; **MEK**, mitogen-activated protein kinase kinase; **mTOR**, mammalian/mechanistic target of rapamycin; **mTORC1**, mTOR complex 1; **PI3K**, phosphatidylinositol 3-kinase; **PTEN**, phosphatase and tensin homolog; **Raf**, rapidly accelerated fibrosarcoma; **Raptor**, regulatory-associated protein of mTOR; **Ras**, rat sarcoma virus homolog; **Rheb**, Ras homolog enriched in brain; **S6K**, ribosomal S6 kinase; **TSC1**, tuberous sclerosis complex subunit 1; **TSC2**, tuberous sclerosis complex subunit 2.

- Clinical use of therapies targeting this pathway is limited by compensatory vertical and horizontal feedback activation loops, which limit antitumor efficacy and lead to drug resistance[1,2]
- Combination therapy strategies employing simultaneous inhibition of multiple PI3K-AKT-mTOR pathway elements may overcome these resistance mechanisms and improve antitumor activity
- *nab*-Sirolimus is an albumin-bound nanoparticle form of the mTOR inhibitor sirolimus
- In preclinical models, *nab*-sirolimus demonstrated significantly greater tumor accumulation, greater mTOR target suppression, and improved antitumor effects, compared with conventional, first-generation mTOR inhibitors[3]
- We analyzed the effects of *nab*-sirolimus combinations in PI3K-mutant breast cancer (BrCa) cells

METHODS

- PI3K-mutated BrCa cell lines (MDA-MD-453, hormone receptor-negative; and MDA-MB-361, hormone receptor-positive) were incubated for 5 days with increasing concentrations of:
 - *nab*-Sirolimus (nanoparticle albumin bound allosteric mTOR inhibitor)
 - The PI3K pathway inhibitors gedatolisib (combination PI3K and mTOR ATP competitive inhibitor)[4] and alpelisib (PI3K alpha specific inhibitor)[5], or
 - The AKT inhibitors miransertib (allosteric pan-AKT inhibitor)[6] and capivasertib (ATP-competitive pan-AKT inhibitor)[7]
- The antiproliferative and cytotoxic effects of single-agent and combination treatment were assessed using cell viability and cell death assays
- Statistical significance of changes in cell viability and cell death were analyzed by two-way analysis of variance
- Cell signaling was analyzed by western blot

RESULTS

nab-Sirolimus in combination with PI3K pathway inhibitors

- In PI3K-mutated BrCa cell lines, the addition of *nab*-sirolimus (20 or 80 nM) to PI3K pathway inhibitors demonstrated both enhanced antiproliferative effects and increased cell death
 - Antiproliferative effects of gedatolisib at low doses (2.5–10 nM) were enhanced by 61%–72% when added in combination with *nab*-sirolimus (20 or 80 nM) to MDA-MB-453 cells ($P < 0.0001$) (**Figure 2A**)
 - Limited cell death (4.2%–9.6%) was observed in MDA-MB-453 cells with low doses (2.5, 5, and 10 nM) of gedatolisib alone; the addition of *nab*-sirolimus (20 or 80 nM) synergistically resulted in high levels of cell death (up to 48.9%; $P \leq 0.0001$ at 5 and 10 nM gedatolisib) (**Figure 2B**)
 - Similar results were observed for alpelisib at 1 µM in combination with *nab*-sirolimus (20 or 80 nM) in MDA-MB-453 cells, with cell proliferation reduced further by ~50% ($P < 0.01$), and cell death rate increased synergistically from 8.8% to >20% ($P < 0.001$) (**Figure 2C**, **D**)
 - Results were similar for both gedatolisib (cell proliferation reduced further by up to 53% at 2.5 nM [$P < 0.0001$], cell death increased synergistically up to 33% [$P < 0.0001$]) and alpelisib (cell proliferation reduced further by >50% [$P < 0.001$], cell death increased synergistically to >20% [$P < 0.0001$]) in combination with *nab*-sirolimus in MDA-MB-361 cells (**Figure 2E**, **F**)

Figure 2. Effect of *nab*-sirolimus in combination with PI3K inhibitors on cell proliferation and cytotoxicity. (**A**) Quantification of proliferation of MDA-MB-453 cells with *nab*-sirolimus (20 or 80 nM) in combination with gedatolisib (2.5–20 nM). (**B**) Quantification of percentage of MDA-MB-453 cell death with *nab*-sirolimus (20 or 80 nM) in combination with gedatolisib (2.5–20 nM). (**C**) Quantification of proliferation of MDA-MB-453 cells with *nab*-sirolimus (20 or 80 nM) in combination with alpelisib (1 µM). (**D**) Quantification of percentage of MDA-MB-453 cell death with *nab*-sirolimus (20 or 80 nM) in combination with alpelisib (1 µM). (**E**) Quantification of proliferation of MDA-MB-361 cells with *nab*-sirolimus (20 or 80 nM) in combination with gedatolisib (2.5–5 nM) or alpelisib (500 nM). (**F**) Quantification of percentage of MDA-MB-361 cell death with *nab*-sirolimus (20 or 80 nM) in combination with gedatolisib (2.5–5 nM) or alpelisib (500 nM).

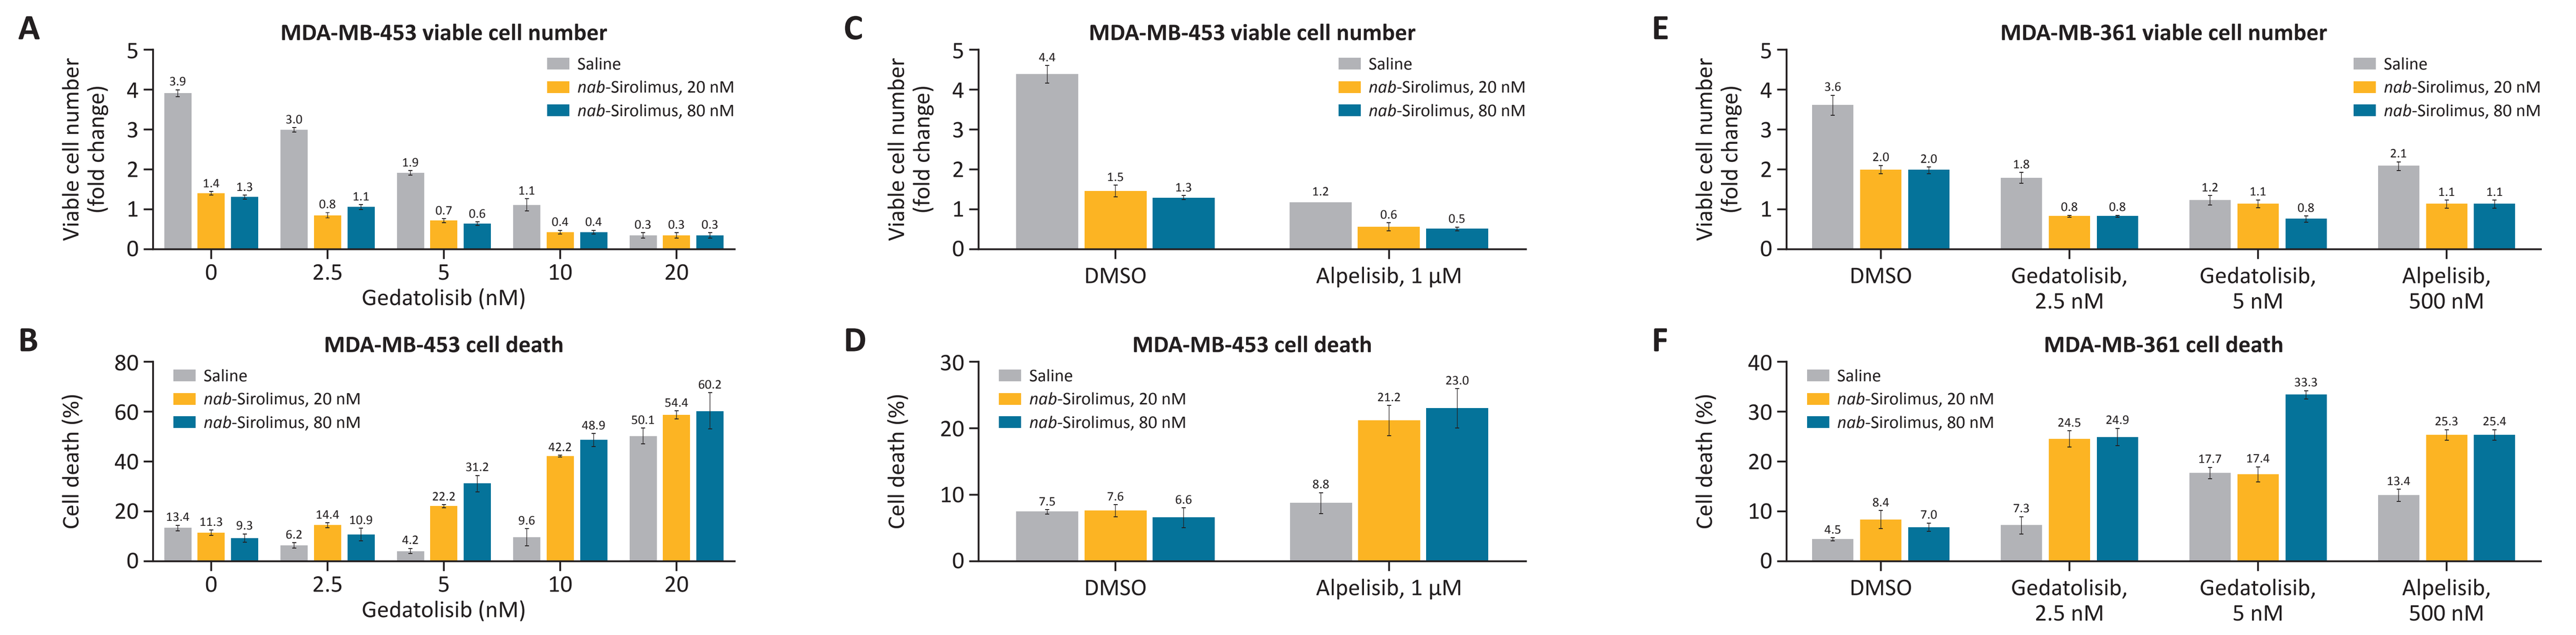

A
MDA-MB-453 viable cell number
Viable cell number (fold change)
Gedatolisib (nM)
Saline
nab-Sirolimus, 20 nM
nab-Sirolimus, 80 nM
B
MDA-MB-453 cell death
Cell death (%)
C
MDA-MB-453 viable cell number
DMSO
Alpelisib, 1 µM
D
MDA-MB-453 cell death
E
MDA-MB-361 viable cell number
Gedatolisib, 2.5 nM
Gedatolisib, 5 nM
Alpelisib, 500 nM
F
MDA-MB-361 cell death

Mean and standard error of the mean are shown. **DMSO**, dimethyl sulfoxide.

nab-Sirolimus in combination with AKT pathway inhibitors

- In PI3K-mutated BrCa cell lines, the addition of *nab*-sirolimus (20 or 80 nM) to AKT inhibitors resulted in both enhanced antiproliferative effects and increased cell death
 - Antiproliferative effects and increased cell death were seen in both MDA-MB-453 ($P < 0.01$ and $P < 0.01$, respectively; **Figure 3A**, **B**) and MDA-MB-361 ($P < 0.05$ and $P < 0.05$, respectively, **Figure 3C**, **D**) cells when *nab*-sirolimus was added to capivasertib (1 or 2 µM)
 - In MDA-MB-361 cells, miransertib (25 or 250 nM) combined with *nab*-sirolimus (80 nM) demonstrated enhanced antiproliferative effects ($P < 0.01$ at 250 nM) and increased cell death ($P < 0.0001$ at 250 nM) (**Figure 3E**, **F**)

Figure 3. Effect of *nab*-sirolimus in combination with AKT inhibitors on cell proliferation and cytotoxicity. (**A**) Quantification of proliferation of MDA-MB-453 cells with *nab*-sirolimus (80 nM) in combination with capivasertib (1 or 2 µM). (**B**) Quantification of percentage of MDA-MB-453 cell death with *nab*-sirolimus (80 nM) in combination with capivasertib (1 or 2 µM). (**C**) Quantification of proliferation of MDA-MB-361 cells with *nab*-sirolimus (80 nM) in combination with capivasertib (1 or 2 µM). (**D**) Quantification of percentage of MDA-MB-361 cell death with *nab*-sirolimus (80 nM) in combination with capivasertib (1 or 2 µM). (**E**) Quantification of proliferation of MDA-MB-361 cells with *nab*-sirolimus (80 nM) in combination with miransertib (25 or 250 nM). (**F**) Quantification of percentage of MDA-MB-361 cell death with *nab*-sirolimus (80 nM) in combination with miransertib (25 or 250 nM).

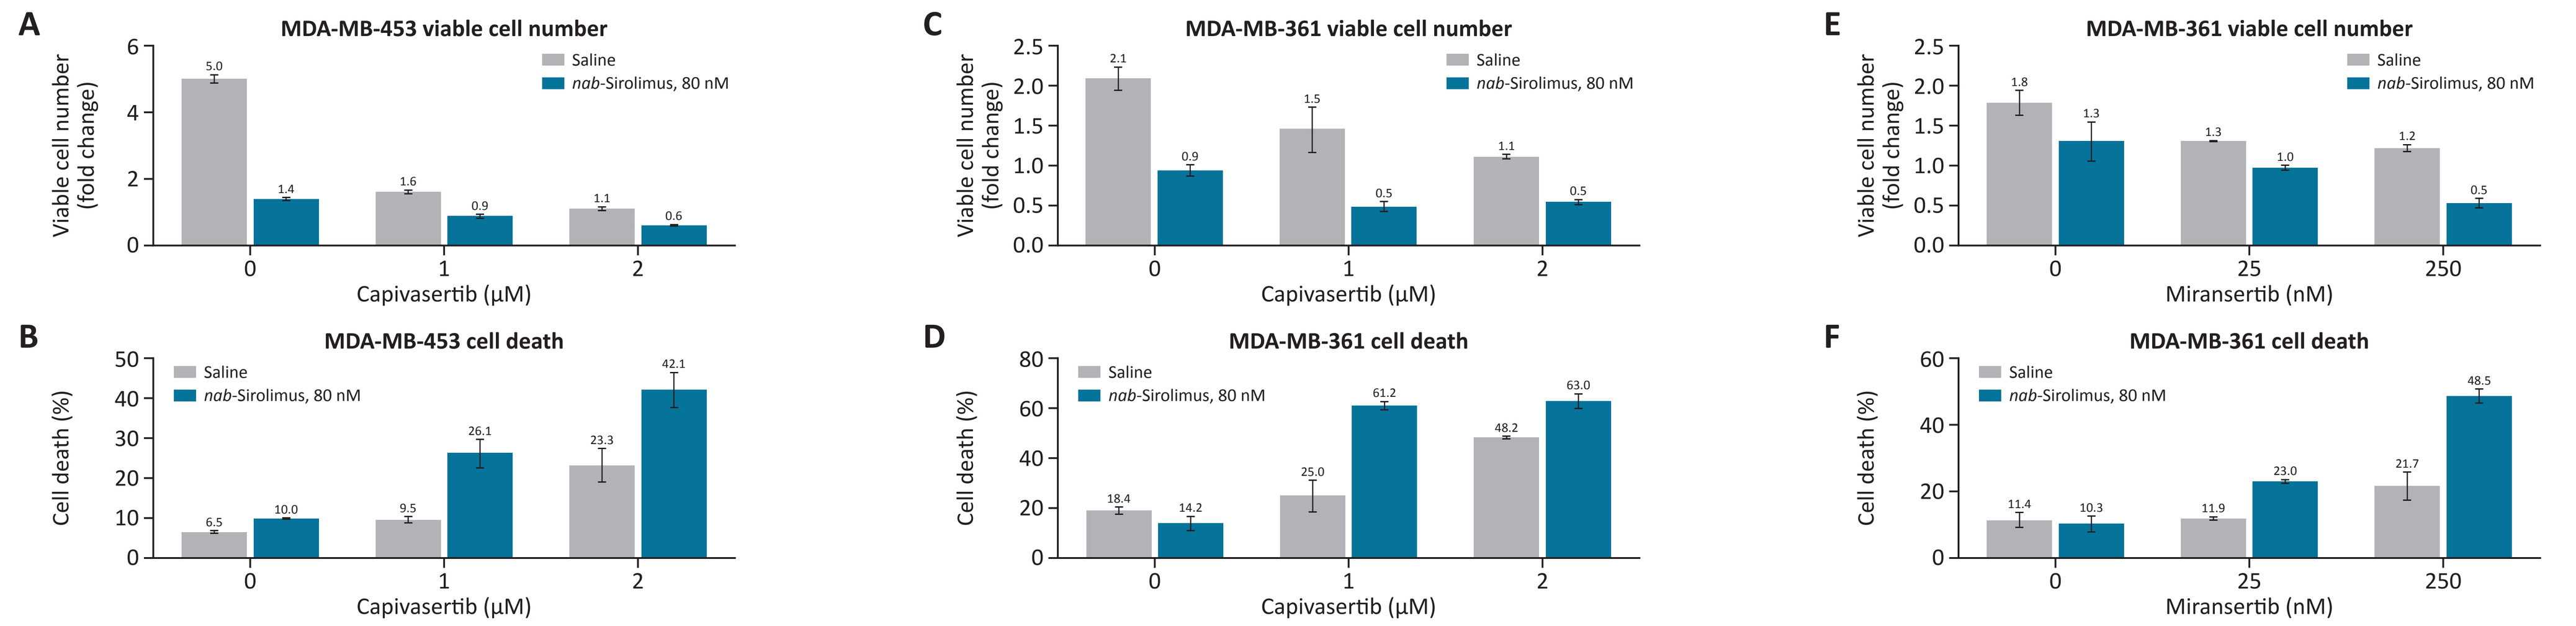

A
MDA-MB-453 viable cell number
Viable cell number (fold change)
Capivasertib (µM)
Saline
nab-Sirolimus, 80 nM
B
MDA-MB-453 cell death
Cell death (%)
C
MDA-MB-361 viable cell number
D
MDA-MB-361 cell death
E
MDA-MB-361 viable cell number
Miransertib (nM)
F
MDA-MB-361 cell death

Mean and standard error of the mean are shown.

nab-Sirolimus in combination with PI3K pathway inhibitors reduces compensatory activation of key signaling molecules

- Western blot analysis was performed on the PI3K-mutated BrCa cell line MDA-MB-453
- Phosphorylation of mTORC1 target S6 kinase and its substrate (S6) was inhibited by the PI3K inhibitors gedatolisib in a dose-dependent manner (**Figure 4A**) or alpelisib alone (**Figure 4B**), and more efficiently by *nab*-sirolimus
- Phosphorylation of the key mTORC1 target 4EBP1 on threonine 70 was upregulated by alpelisib alone by 301% (**Figure 4B**); gedatosolib (PI3K and mTOR ATP competitive inhibitor) still caused upregulation by 128%–372%; however, the combination of gedatosolib and *nab*-sirolimus (allosteric mTOR inhibitor) decreased phosphorylation of 4EBP1 even further below that of *nab*-sirolimus on its own
- The phospho-AKT level was constitutively high in PI3K-mutant MDA-MB-453 cells and was further increased (up to 250%) by *nab*-sirolimus through a negative feedback loop, which can be completely inhibited by combination with the PI3K inhibitors gedatolisib (**Figure 4A**) and alpelisib (**Figure 4B**)
- The compensatory feedback activation loops observed with single-agent PI3K inhibitors and *nab*-sirolimus were both reversed by combining these agents, as indicated by dramatically decreased p4EBP1 and pAKT when the agents were combined
- Western blot results correlated with cell proliferation and cell death results, demonstrating the reciprocal benefits of combining *nab*-sirolimus with PI3K inhibitors

Figure 4. Western blot analysis demonstrating PI3K-AKT-mTOR pathway crosstalk in MDA-MB-453 BrCa cells. Western blots showing phospho-P70 S6 kinase (mTORC1 target) and P70 S6 kinase total, phospho-S6 (S6 kinase target) and S6 total, phospho-4EBP1 (mTORC1 target) and 4EBP1 total, phospho-AKT (activated with mTORC1 inhibition) and total AKT, cPARP (apoptotic marker) and GAPDH (control) with *nab*-sirolimus (20 or 80 nM) in combination with (**A**) gedatolisib (2.5–20 nM) or (**B**) alpelisib (1 µM).

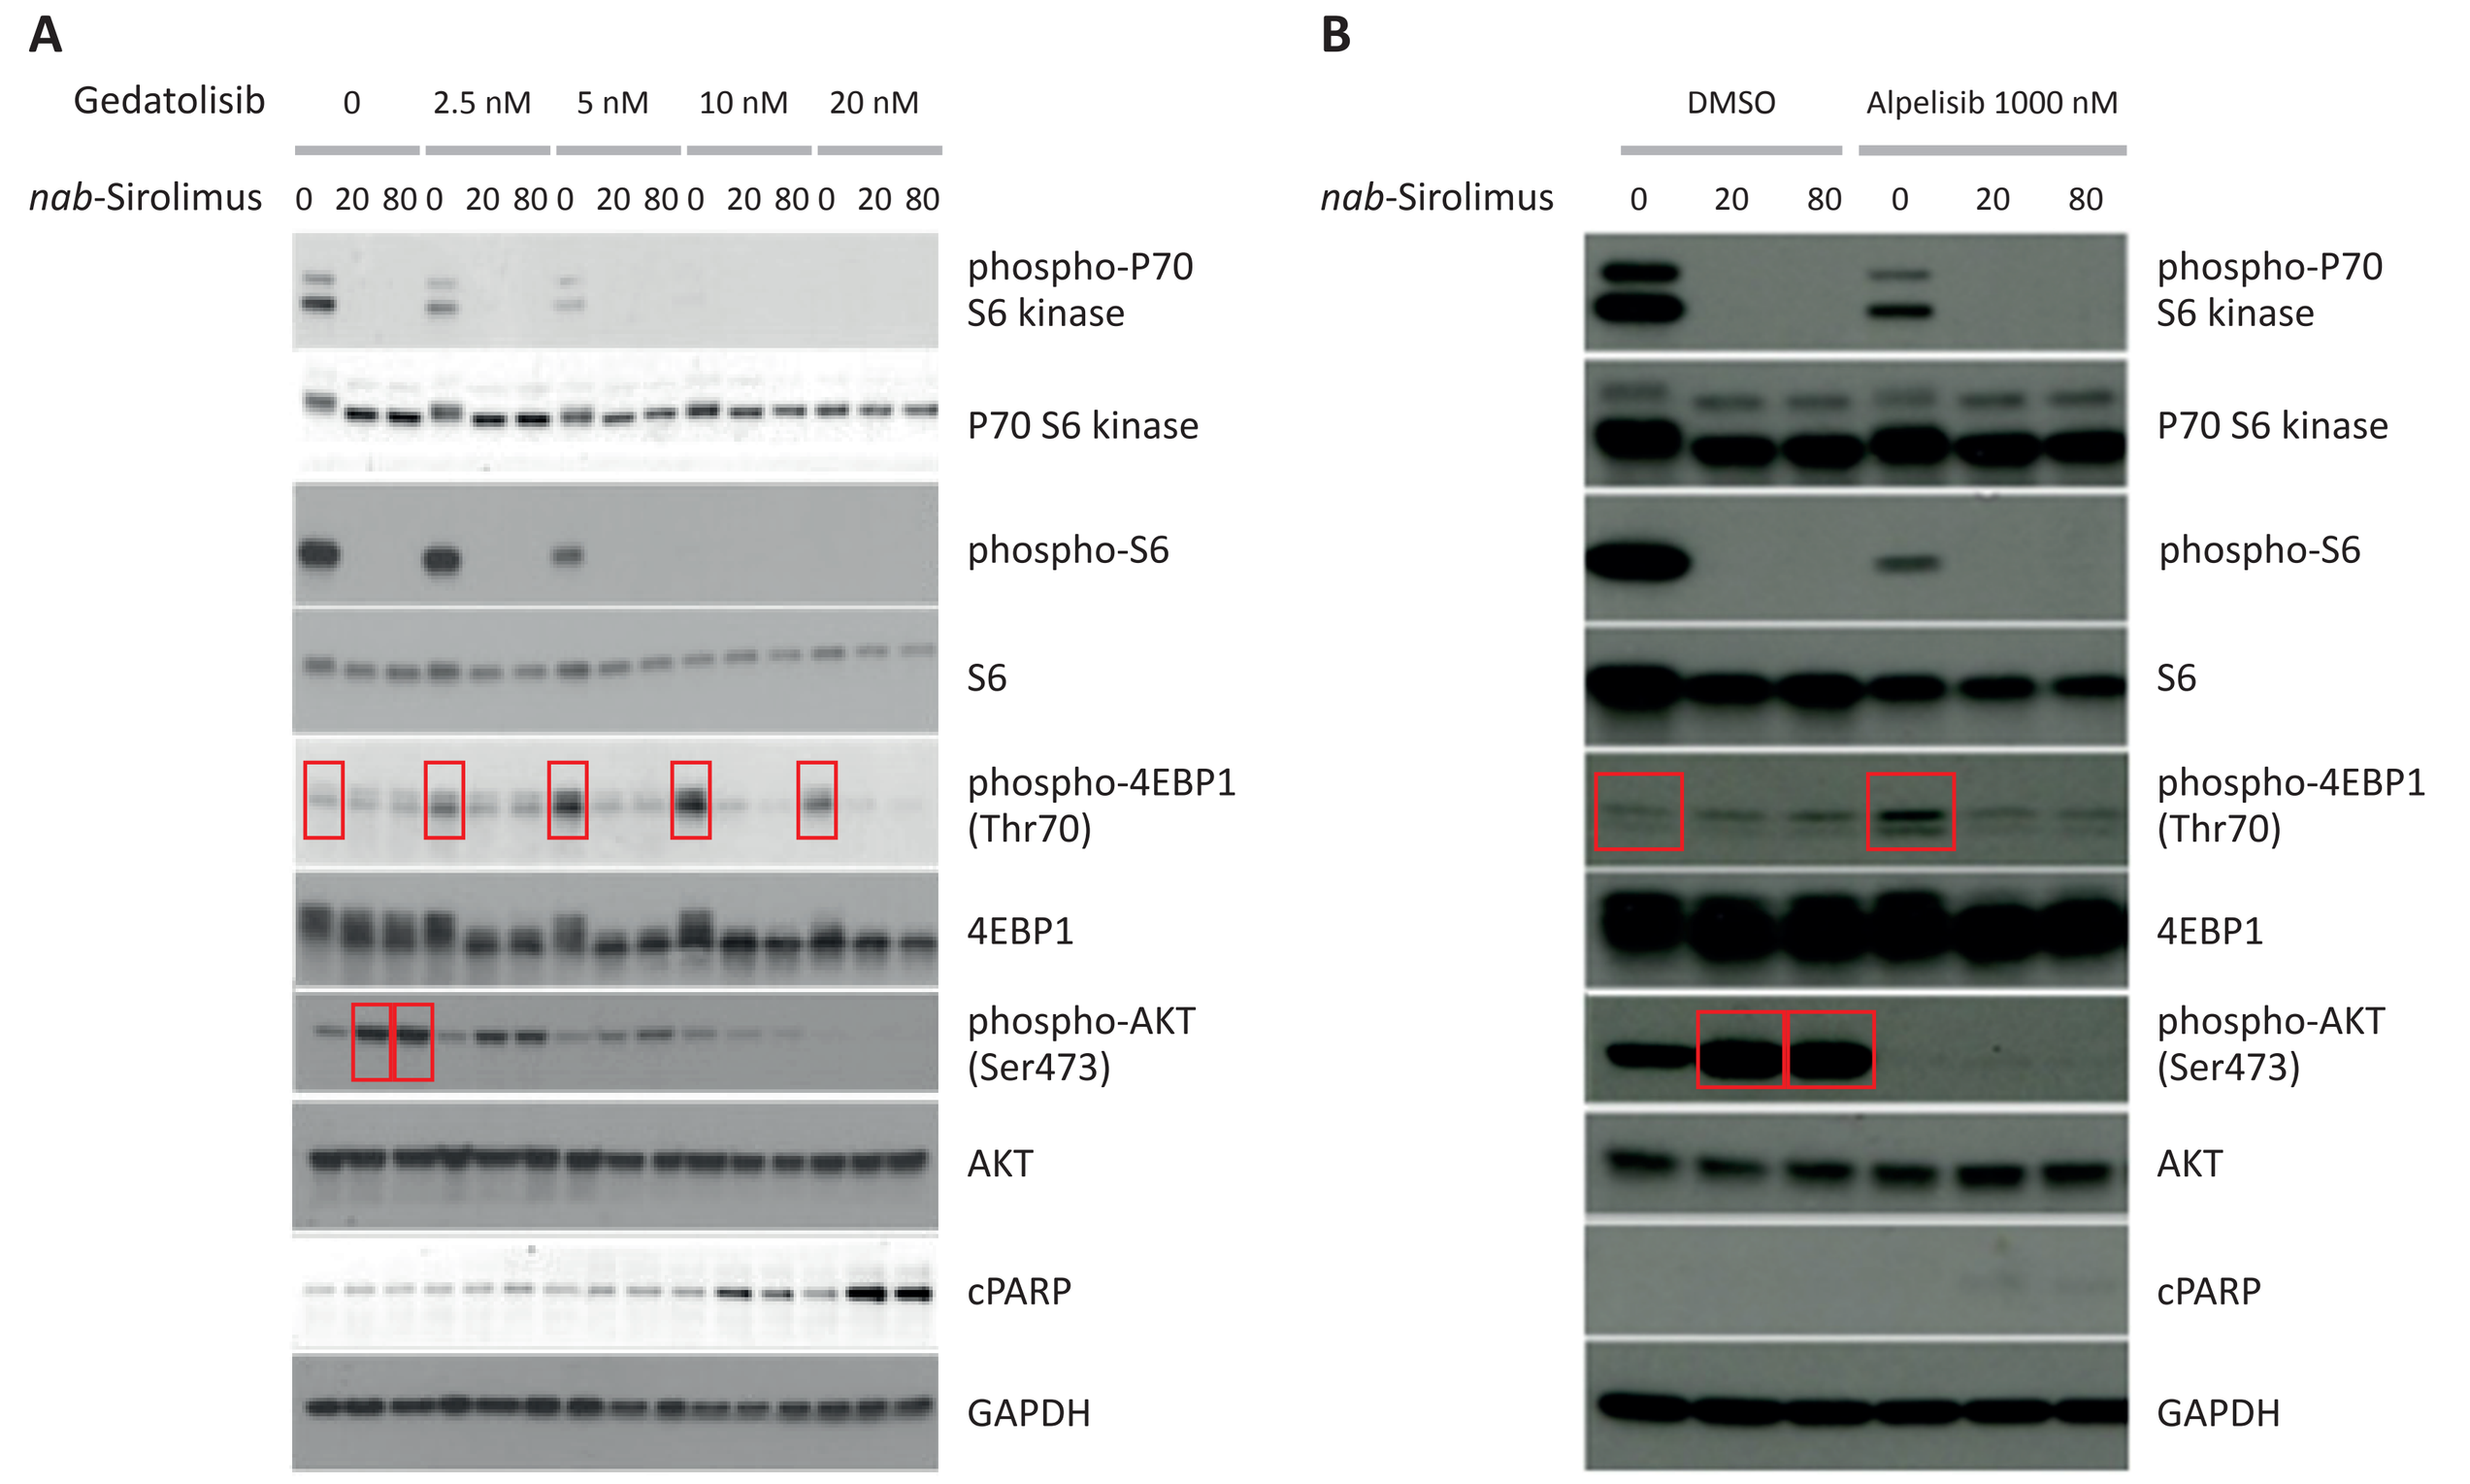

A
Gedatolisib
0
2.5 nM
5 nM
10 nM
20 nM
nab-Sirolimus
0 20 80 0 20 80 0 20 80 0 20 80 0 20 80
phospho-P70 S6 kinase
P70 S6 kinase
phospho-S6
S6
phospho-4EBP1 (Thr70)
4EBP1
phospho-AKT (Ser473)
AKT
cPARP
GAPDH
B
DMSO
Alpelisib 1000 nM
nab-Sirolimus
0 20 80 0 20 80

4EBP1, eukaryotic initiation factor 4E-binding protein 1; **AKT**, protein kinase B; **cPARP**, cleaved poly (ADP-ribose) polymerase; **DMSO**, dimethyl sulfoxide; **GAPDH**, glyceraldehyde 3-phosphate dehydrogenase; **mTORC1**, mammalian/mechanistic target of rapamycin complex 1; **P70 S6 kinase**, ribosomal protein S6 kinase beta-1; **phospho**, phosphorylated; **PI3K**, phosphoinositide 3-kinase; **Ser**, serine; **Thr**, threonine.

Acknowledgments & Disclosures

Medical writing and editorial assistance were provided by Heather Caballes, PhD, of Twist Medical, and were funded by Aadi Bioscience, Inc.

SW, **KNM**, and **IV**: Report no disclosures. **SH**, **MZ**, **AK**, and **BM**: Employment and stockownership in Aadi Biosciences.

Correspondence: MedInfo@AadiBio.com

References

1. Peng Y, et al. *Front Oncol.* 2022;12:819128.
2. Yang J, et al. *Mol Cancer.* 2019;18:26.
3. Hou S, et al. *Cancer Res.* 2019;79 (13_Supplement):348.
4. Dehnhardt CM, et al. *Bioorg Med Chem Lett.* 2011;21:4773–8.
5. Andre F, et al. *N Eng J Med.* 2019;380:1929–40.
6. Biesecker LG, et al. *Cold Spring Harb Mol Case Stud.* 2020;6:a004549.
7. Andrikopoulou A, et al. *Breast.* 2022;63:157–67.

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from AACR-NCI-EORTC or the author of the poster